Molluscum Contagiosum: Persistent Burden, Unmet Need, and a Mechanism–Treatment Mismatch Despite Recent Therapeutic Advances

Rich Panico[1], Sunil S Dhawan[2]
[1] Precefi, Inc., Port Jefferson, United States, [2] Center for Dermatology Clinical Research, Fremont, United States

Background:

Molluscum contagiosum (MC) is a contagious poxvirus infection caused by molluscum contagiosum virus (MCV; genus *Molluscipoxvirus*). A CDC-authored study reported a historical annual U.S. outpatient visit rate of ~22 per 10,000 persons;[1] with most cases not reaching medical attention, true incidence is considerably higher.[2] MC predominantly affects children aged 1–10 years. In 2023–2024, cantharidin 0.7% (YCANTH) and berdazimer gel 10.3% (Zelsuvmi) received FDA approval as the first dedicated MC therapies. Despite these advances, many cases remain untreated, and the clinical and psychosocial burden persists.[3] Less attention has been paid to how mechanistic limitations of available therapies may themselves shape observed epidemiology and care-seeking behavior.

Objectives:

To characterize the persistent unmet need and clinical, psychosocial, and behavioral burden of MC despite recent FDA approvals; evaluate the alignment of current therapies with viral pathophysiology; and examine how therapeutic limitations may shape patient care-seeking.

Methods:

A targeted review was conducted of epidemiologic data, patient-reported outcomes, and pivotal phase 3 trials of cantharidin 0.7% and berdazimer gel 10.3%.[4,5,6] Outcomes were evaluated alongside mechanism of action, tolerability, and mode of administration. Therapeutic inflection points in onychomycosis and herpes zoster were referenced as comparators.

Results:

MC lesions may persist for 8–18 months or longer, with patient-reported durations of up to 2 years in trial populations.[3,6,9] Household transmission rates approach 41%, extending burden across siblings and caregivers.[9] Visible, exposed-area lesions contribute to self-consciousness, activity restriction, and social exclusion in affected children.[6] Cantharidin demonstrated complete clearance in 46–54% of patients in phase 3 trials but produced local skin reactions in ~97% and required in-office administration across multiple visits.[4] Berdazimer gel enables at-home application but achieved complete clearance in roughly one-third of patients at 12 weeks.[5]

Mechanistically, MCV replicates exclusively within keratinocytes and employs immune-evasion strategies that limit host clearance.[12] Cantharidin is a vesicant, and berdazimer is a nitric oxide-releasing agent whose mechanism of action in treating MC is unknown. [13] Neither therapy has demonstrated direct, clinically confirmed antiviral activity against MCV.

In a 2009 U.S. survey, 41.6% of healthcare providers selected reassurance that molluscum contagiosum would resolve when asked what advice they would give patients.[7] For comparison, in onychomycosis, where approximately half of affected patients historically failed to seek medical advice,[8] oral terbinafine offered improved cure rates and tolerability, repositioning the condition from one largely tolerated without intervention to one actively managed. Analogous inflection points following effective herpes zoster and herpes labialis therapies suggest that observed MC epidemiology may partially reflect treatment-driven care avoidance rather than true disease infrequency.

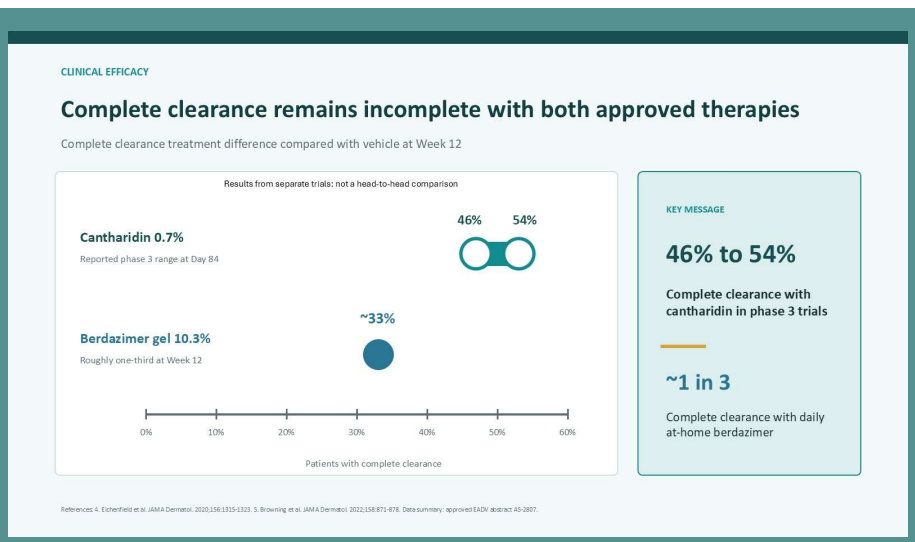
CLINICAL EFFICACY
Complete clearance remains incomplete with both approved therapies
Complete clearance treatment difference compared with vehicle at Week 12
Results from separate trials: not a head-to-head comparison
Cantharidin 0.7%
Reported phase 3 range at Day 84
46%
54%
Berdazimer gel 10.3%
Roughly one-third at Week 12
~33%
0%
10%
20%
30%
40%
50%
60%
Patients with complete clearance
KEY MESSAGE
46% to 54%
Complete clearance with cantharidin in phase 3 trials
~1 in 3
Complete clearance with daily at-home berdazimer
References 4. Eichenfield et al. JAMA Dermatol. 2020;156:1315-1323. 5. Browning et al. JAMA Dermatol. 2022;158:871-878. Data summary: approved EADV abstract A5-2807.

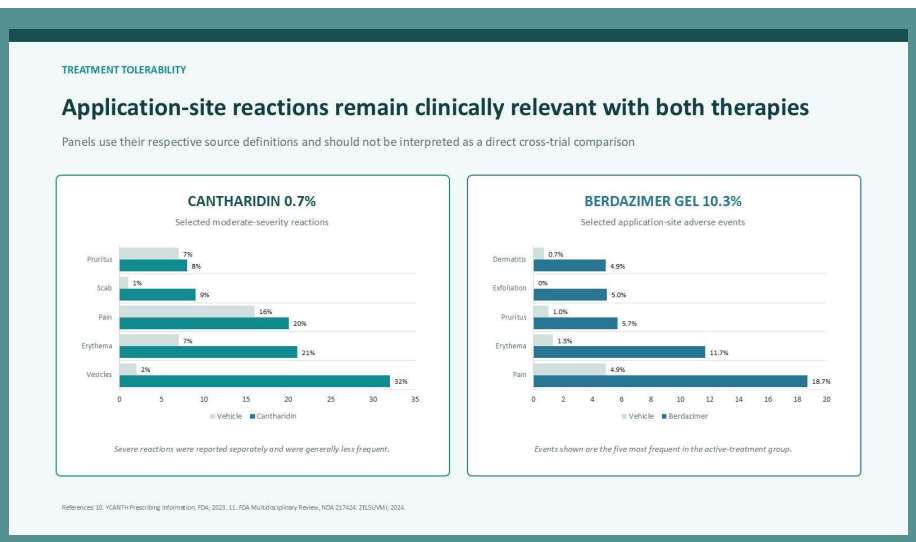
TREATMENT TOLERABILITY
Application-site reactions remain clinically relevant with both therapies
Panels use their respective source definitions and should not be interpreted as a direct cross-trial comparison
CANTHARIDIN 0.7%
Selected moderate-severity reactions
Pruritus
Scab
Pain
Erythema
Vesicles
7%
8%
1%
9%
16%
20%
7%
21%
2%
32%
Vehicle
Cantharidin
Severe reactions were reported separately and were generally less frequent.
BERDAZIMER GEL 10.3%
Selected application-site adverse events
Dermatitis
Exfoliation
Pruritus
Erythema
Pain
0.7%
4.9%
0%
5.0%
1.0%
5.7%
1.3%
11.7%
4.9%
18.7%
Vehicle
Berdazimer
Events shown are the five most frequent in the active-treatment group.

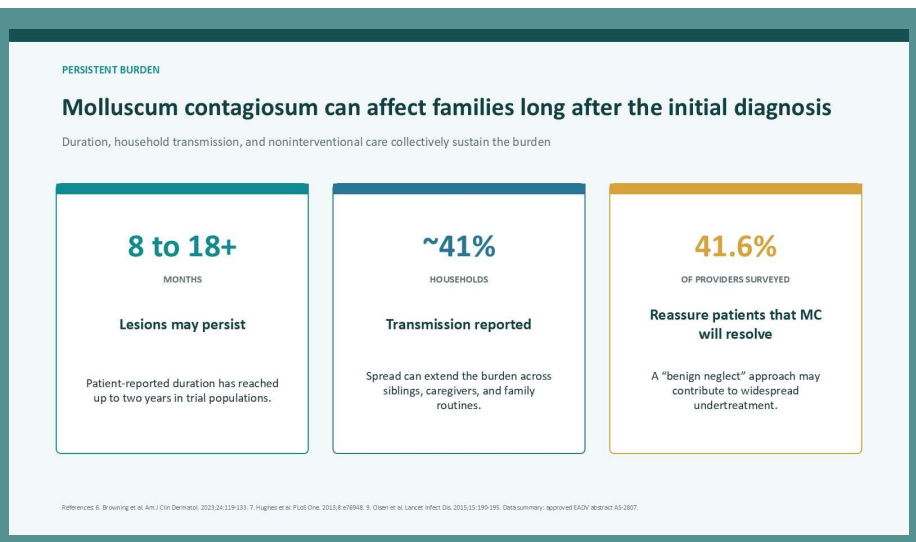
PERSISTENT BURDEN
Molluscum contagiosum can affect families long after the initial diagnosis
Duration, household transmission, and noninterventional care collectively sustain the burden
8 to 18+
MONTHS
Lesions may persist
Patient-reported duration has reached up to two years in trial populations.
~41%
HOUSEHOLDS
Transmission reported
Spread can extend the burden across siblings, caregivers, and family routines.
41.6%
OF PROVIDERS SURVEYED
Reassure patients that MC will resolve
A "benign neglect" approach may contribute to widespread undertreatment.

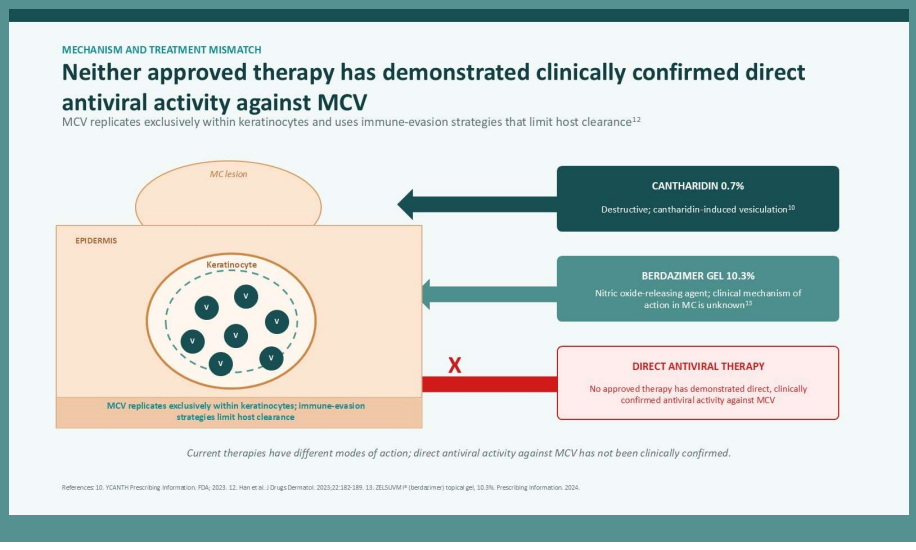
MECHANISM AND TREATMENT MISMATCH
Neither approved therapy has demonstrated clinically confirmed direct antiviral activity against MCV
MCV replicates exclusively within keratinocytes and uses immune-evasion strategies that limit host clearance12
MC lesion
EPIDERMIS
Keratinocyte
MCV replicates exclusively within keratinocytes; immune-evasion strategies limit host clearance
CANTHARIDIN 0.7%
Destructive; cantharidin-induced vesiculation10
BERDAZIMER GEL 10.3%
Nitric oxide-releasing agent; clinical mechanism of action in MC is unknown13
X
DIRECT ANTIVIRAL THERAPY
No approved therapy has demonstrated direct, clinically confirmed antiviral activity against MCV
Current therapies have different modes of action; direct antiviral activity against MCV has not been clinically confirmed.

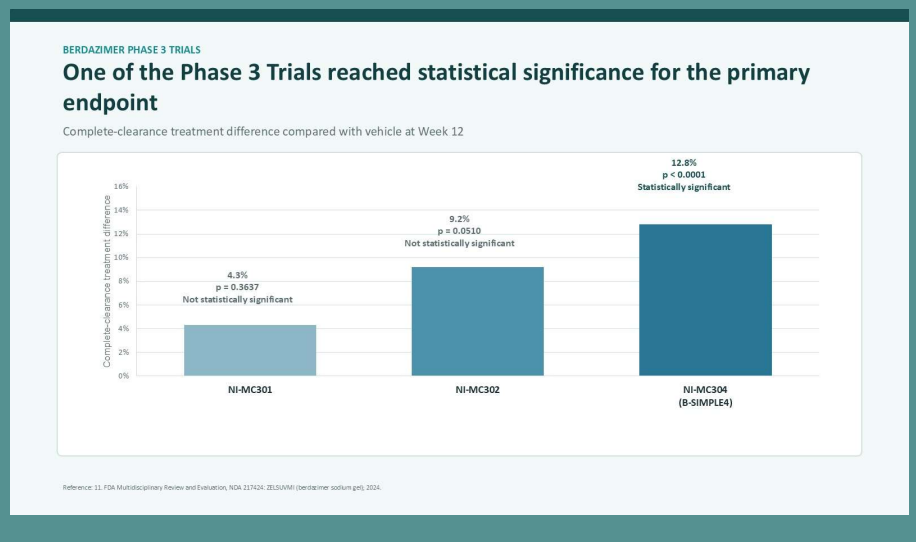
BERDAZIMER PHASE 3 TRIALS
One of the Phase 3 Trials reached statistical significance for the primary endpoint
Complete-clearance treatment difference compared with vehicle at Week 12
Complete-clearance treatment difference
4.3%
p = 0.3637
Not statistically significant
9.2%
p = 0.0510
Not statistically significant
12.8%
p < 0.0001
Statistically significant
NI-MC301
NI-MC302
NI-MC304
(B-SIMPLE4)
Reference: 11. FDA Multidisciplinary Review and Evaluation, NDA 217424: ZELSUVMI (berdazimer sodium gel). 2024.

Conclusion:

Despite recent FDA approvals, MC continues to carry substantial clinical and psychosocial burden. Current therapies do not consistently achieve complete lesion clearance or meet patient and caregiver needs for tolerability and convenience. The resulting mechanism–treatment mismatch may function not only as a clinical limitation but as a driver of suppressed care-seeking. Therapies that are both directly antiviral and meaningfully better tolerated may improve clinical outcomes and expand appropriate treatment engagement within a large, underserved patient population.

[1]Reynolds MG, Holman RC, Yorita Christensen KL, Cheek JE, Damon IK. The incidence of molluscum contagiosum among American Indians and Alaska Natives. PLoS One. 2009;4(4):e5255.
[2]McCollum AM, Holman RC, Hughes CM, et al. Molluscum contagiosum in a pediatric American Indian population: incidence and risk factors. PLoS One. 2014;9(7):e103419.
[3]Centers for Disease Control and Prevention. Clinical Overview of Molluscum Contagiosum. Updated April 15, 2025. Accessed May 6, 2026. https://www.cdc.gov/molluscum-contagiosum/hcp/clinical-overview/index.html.
[4]Eichenfield LF, McFalda W, Brabec B, et al. Safety and efficacy of VP-102, a proprietary drug-device combination product containing cantharidin, 0.7% (w/v), in children and adults with molluscum contagiosum: two phase 3 randomized clinical trials. JAMA Dermatol. 2020;156(12):1315-1323.
[5]Browning JC, Enloe C, Cartwright M, et al. Efficacy and safety of topical nitric oxide-releasing berdazimer gel in patients with molluscum contagiosum: a phase 3 randomized clinical trial. JAMA Dermatol. 2022;158(8):871-878.
[6]Browning JC, Cartwright M, Thorla I, et al. A patient-centered perspective of molluscum contagiosum as reported by B-SIMPLE4 clinical trial patients and caregivers: global impression of change and exit interview substudy results. Am J Clin Dermatol. 2023;24(1):119-133.
[7]Hughes CM, Damon IK, Reynolds MG. Understanding U.S. healthcare providers' practices and experiences with molluscum contagiosum. PLoS One. 2013;8(10):e76948.
[8]Roberts DT. Onychomycosis: current treatment and future challenges. Br J Dermatol. 1999;141(Suppl 56):1-4.
[9]Olsen JR, Gallacher J, Finlay AY, Piguet V, Francis NA. Time to resolution and effect on quality of life of molluscum contagiosum in children in the UK: a prospective community cohort study. Lancet Infect Dis. 2015;15(2):190-195.
[10]U.S. Food and Drug Administration. YCANTH (cantharidin) topical solution, 0.7%. Prescribing Information. 2023. Accessed August 11, 2026. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/212905s000lbl.pdf.
[11]U.S. Food and Drug Administration. NDA/BLA Multi-Disciplinary Review and Evaluation, NDA 217424: ZELSUVMI (berdazimer sodium gel). Review completion date: January 4, 2024. Accessed August 11, 2026. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2024/217424Orig1s000MultidisciplineR.pdf.
[12]Han H, Smythe C, Yousefian F, Berman B. Molluscum contagiosum virus evasion of immune surveillance: a review. J Drugs Dermatol. 2023;22(2):182-189. doi:10.36849/JDD.7230.
[13]U.S. Food and Drug Administration. *ZELSUVMI (berdazimer) topical gel, 10.3%. Prescribing Information.* 2024. Accessed August 11, 2026. https://www.accessdata.fda.gov/drugsatfda_docs/label/2024/217424s000lbl.pdf.

Scan me!